GRIP
GLOBAL RESPIRATORY INFECTION PARTNERSHIP

RESPONSIBILITY TREATS BEST
STOP ANTIBIOTIC OVERUSE

P2248

UNDERSTANDING ATTITUDES AND PERCEPTIONS OF YOUNGER ADULTS IN RELATION TO ANTIBIOTIC USE FOR RESPIRATORY ILLNESS: RESULTS FROM THE SORE THROAT AND ANTIBIOTIC RESISTANCE (STAR) STUDY

Sabiha Essack,[1] Sergio Caretta-Barradas,[2] Antonio Pignatari,[3] Khalid Eljaaly,[4] Wirat Tongrod,[5] Adrian Shephard,[6] Alike van der Velden[7]

[1]Antimicrobial Research Unit, College of Health Sciences, University of KwaZulu-Natal, Durban, South Africa; [2]Respiratory Disease Centre, Mexico City, Mexico; [3]Paulista School of Medicine (EPM), Federal University of São Paulo (UNIFESP), Brazil; [4]Faculty of Pharmacy, King Abdulaziz University, Jeddah, Saudi Arabia; [5]Faculty of Pharmaceutical Sciences, Huachiew Chalermprakiet University, Thailand; [6]Reckitt Benckiser Healthcare International Ltd, Slough, Berkshire, UK; [7]University Medical Center Utrecht, The Netherlands

Khalid Eljaaly – Presenting Author

INTRODUCTION

- Overuse and misuse of antibiotics are the main drivers of antimicrobial resistance (AMR)[1]
- Inappropriate use of antibiotics is likely to be influenced, amongst other things, by patients' misunderstanding of how antibiotics work[2-6]
- The World Health Organization (WHO) is leading initiatives to optimise the use of antibiotics and increase public awareness and understanding of antibiotics and AMR[7]
- In order to tailor communication and track the effectiveness of public awareness campaigns, it is important to understand how misconceptions about antibiotics vary across age groups and geographical locations through targeted research
- The insights gained from such research will help to generate targeted messaging and materials to drive behaviour change and reduce inappropriate antibiotic use and AMR

AIM

- To understand the attitudes and perceptions of adults in relation to antibiotic use for respiratory illness across Brazil, Mexico, The Philippines, Saudi Arabia, Thailand and South Africa, and evaluate any differences within younger populations, specifically 25–34-year-olds

METHODS

Study design

- An observational, questionnaire-based study was conducted across 12 countries from the WHO regions (Americas [Brazil, Mexico], Europe [Germany, Spain, Italy, Poland, Romania, the United Kingdom], Western Pacific [The Philippines], Southeast Asia [Thailand], Eastern Mediterranean [Saudi Arabia] and Africa [South Africa]). Here we present the findings from the six countries within the five WHO regions outside Europe (n=6000)
 - Results from Europe have been presented at the European Congress of Clinical Microbiology and Infectious Diseases (ECCMID) in April 2023[8]
- Online 15–20-minute interviews were conducted in May 2022
- The questionnaire explored respondent experiences of respiratory illness, including sore throat and other symptoms, antibiotic use and attitudes to and perceptions of antibiotics
- For questions on attitudes and perceptions to antibiotics and perceptions on when antibiotics should be stopped, respondents were asked to give their response using a 7-point Likert scale:
 - Agree strongly; agree somewhat; neither agree nor disagree; disagree somewhat; disagree strongly; would rather not say; don't know. The results presented represent those indicating 'agree strongly' and 'agree somewhat'
- For the question on how much information had been seen in the last 6 months about the unnecessary use of antibiotics, respondents were asked to choose from one of the following options:
 - A great deal; a fair amount; a small amount; have not seen any; would rather not say; don't know. The results presented represent those indicating 'a small amount' and 'have not seen any'

Inclusion criteria

- Healthy adults aged 18–64 years (recruited from a research panel)
- From any of the six countries within the five WHO regions listed above (n=1000 per country)
- Had experienced and treated respiratory symptoms (maximum 15% per country caused by COVID-19) in the past 6 months

Exclusion criteria

- Had not experienced respiratory symptoms that they had treated in the past 6 months
- Indicated that they would 'rather not say' or 'did not know' what the underlying respiratory condition for their symptom was
- Symptoms were not part of a respiratory condition or part of a long-term illness

RESULTS

Study population

- In total, 18,564 people were screened and 12,000 were eligible and completed the questionnaire
 - 50% (n=6000) were from the six countries within the five WHO regions, and of these, 18% (n=1074) were aged 18–24 years, 30% (n=1816) were aged 25–34 years, 26% (n=1583) were aged 35–44 years, 17% (n=1008) were aged 45–54 years and 9% (n=519) were aged 55–64 years

Attitudes and perceptions around antibiotics

- Of all respondents, 65% (n=3929) thought that they were quite knowledgeable about how antibiotics work on respiratory symptoms; in 25–34-year-olds this response ranged from 54% (Brazil [n=150]) to 74% (The Philippines [n=233]). However, answers to further questions indicated substantial misunderstanding overall and specifically among 25–34-year-olds (**Table 1**)
 - 65% (n=3886) agreed that antibiotics kill viruses, 73% (n=4360) believed that antibiotics were effective for sore throat, and 65% (n=3892) agreed antibiotics relieve pain
 - A slightly higher proportion of 25–34-year-olds agreed with these statements, demonstrating a lower understanding (68% [n=1226], 74% [n=1344] and 68% [n=1220], respectively)
 - Almost half of all respondents (48% [n=2857]) felt they do not have enough knowledge on how to treat respiratory symptoms without antibiotics; similar results were observed in 25–34-year-olds (49% [n=881])
 - Overall, 40% (n=2406) would feel anxious about being treated for respiratory symptoms without antibiotics; in 25–34-year-olds this response ranged from 33% (South Africa [n=105]) to 55% (Thailand [n=132])

Table 1. Attitudes and perceptions around antibiotics from study population (n=6000)

Attitude/perception	Six countries within five WHO regions (18–64-year-olds) n=6000	25–34-year-olds (n=1816)						
		Total n=1816	Brazil n=280	Mexico n=297	The Philippines n=316	Saudi Arabia n=369	South Africa n=315	Thailand n=239
Antibiotics...								
kill viruses	65%	68%	56%	62%	79%	66%	68%	75%
are effective for sore throat	73%	74%	80%	69%	81%	71%	67%	77%
relieve pain	65%	68%	70%	61%	71%	65%	62%	76%
are effective for colds and the flu	70%	73%	61%	66%	83%	71%	77%	77%
quickly relieve symptoms	74%	74%	72%	73%	81%	67%	74%	82%
should be available on request from the pharmacy	66%	68%	60%	71%	75%	61%	70%	69%
I do not have enough knowledge to know how to treat respiratory symptoms without antibiotics	48%	49%	48%	49%	48%	47%	47%	54%
I would feel anxious about being treated for respiratory symptoms without antibiotics	40%	44%	34%	44%	50%	48%	33%	55%
I am quite knowledgeable about how antibiotics work on respiratory symptoms/ conditions	65%	66%	54%	66%	74%	72%	64%	64%

Percentages highlighted in red indicate a higher percentage compared with 18–64-year-olds for five WHO regions (n=6000)

Attitudes on stopping antibiotics

- There were misconceptions around when antibiotics should be stopped
 - **'You should stop taking antibiotics as quickly as possible'**

 Of all respondents, 40% (n=2413) agreed with this statement; in 25–34-year-olds, this response ranged from 29% (South Africa [n=90]) to 52% (Thailand [n=123])
 - **'You should stop taking antibiotics if you don't get better in a couple of days'**

 Of all respondents, 48% (n=2859) agreed with this statement; in 25–34-year-olds, this response ranged from 37% (South Africa [n=116]) to 61% (Thailand [n=145])
 - **'You should stop taking antibiotics when you feel better'**

 Of all respondents, 47% (n=2848) agreed with this statement; in 25–34-year-olds, this response ranged from 42% (South Africa [n=132]) to 65% (Thailand [n=156])

Attitudes regarding leftover antibiotics

- Across the six countries within five WHO regions there were varied opinions across the age groups on whether leftover antibiotics should be kept for future use
 - The highest proportion of respondents who agreed with the statement '*You should keep leftover antibiotics for the next time you or your family get sick*' was found in 25–34-year-olds and the lowest proportion was found in 55–64-year-olds (**Figure 1**)

Figure 1. Proportion of patients in each age group across the six countries within five WHO regions who agreed that leftover antibiotics should be kept for future use*

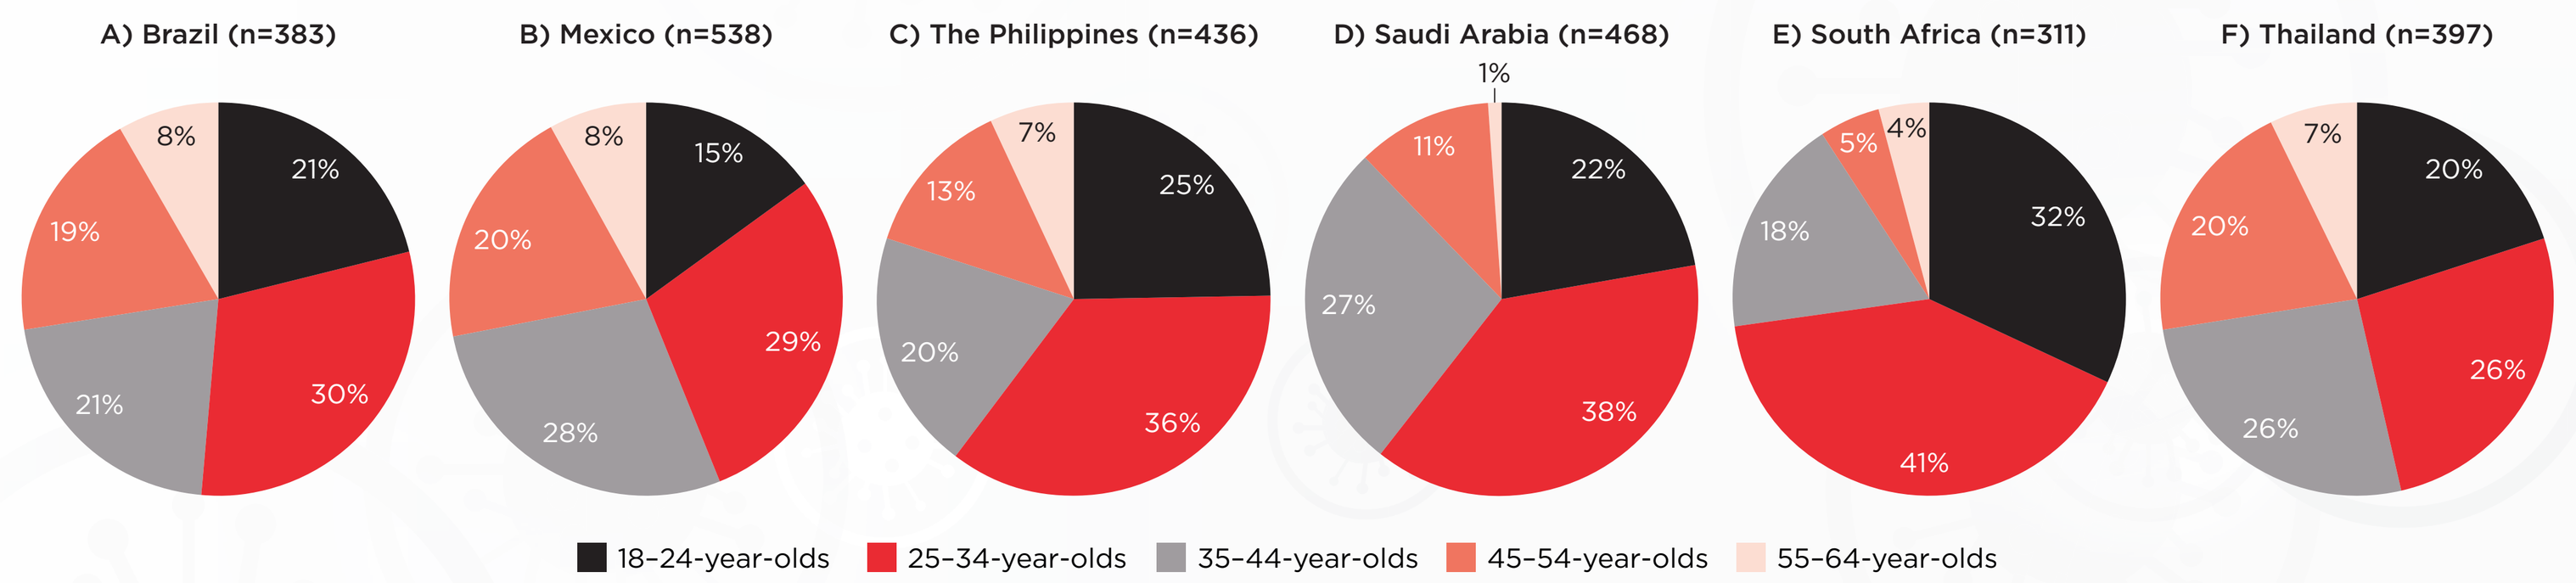

A) Brazil (n=383)
8%
21%
19%
21%
30%
B) Mexico (n=538)
8%
15%
20%
29%
28%
C) The Philippines (n=436)
7%
25%
13%
20%
36%
D) Saudi Arabia (n=468)
1%
11%
22%
27%
38%
E) South Africa (n=311)
5%
4%
18%
32%
41%
F) Thailand (n=397)
7%
20%
20%
26%
26%
18–24-year-olds
25–34-year-olds
35–44-year-olds
45–54-year-olds
55–64-year-olds

*Percentages may not total 100 due to rounding

Information on the unnecessary use of antibiotics

- When asked how much information they have seen about the unnecessary use of antibiotics, 51% (n=3035) of respondents reported they had not seen any or had only seen a small amount of information (**Figure 2**)
- There was wide variation between countries, specifically in the younger population (25–34-year-olds), regarding the amount of information they had seen in the last 6 months about the unnecessary use of antibiotics (**Table 2**)
 - Respondents from Brazil and Mexico were least likely to have seen any information about the unnecessary use of antibiotics

Table 2. Amount of information seen about the unnecessary use of antibiotics in 25–34-year-olds across the six countries within five WHO regions

25–34-year-olds (n=1816)						
Total	**Brazil**	**Mexico**	**The Philippines**	**Saudi Arabia**	**South Africa**	**Thailand**
n=1816	n=280	n=297	n=316	n=369	n=315	n=239
853 (47%)	173 (62%)	184 (62%)	114 (36%)	97 (26%)	187 (59%)	98 (41%)

Figure 2. Amount of information seen about the unnecessary use of antibiotics from the overall study population within five WHO regions (n=6000)*

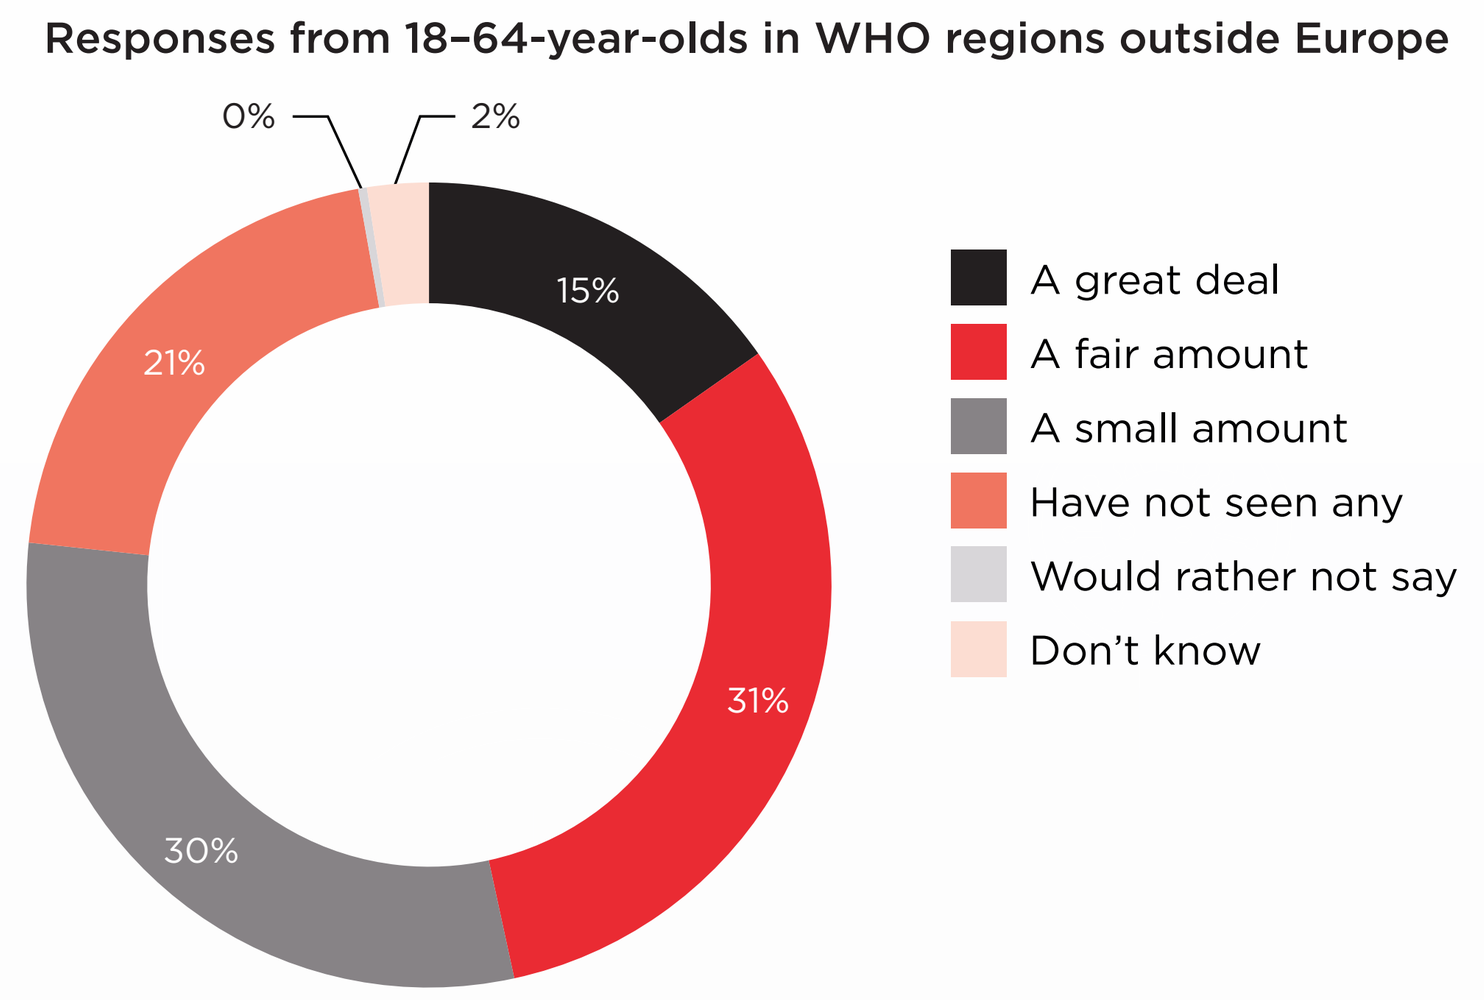

Responses from 18–64-year-olds in WHO regions outside Europe
0%
2%
15%
21%
31%
30%
A great deal
A fair amount
A small amount
Have not seen any
Would rather not say
Don't know

*Percentages may not total 100 due to rounding

CONCLUSIONS

- Despite global efforts to improve public awareness and understanding of AMR, this study demonstrates that there remain gaps in public knowledge regarding the appropriate use of antibiotics
- Conveying the correct messages to the public in relation to appropriate antibiotic use is essential and would help to support a reduction in antibiotic overuse and tackle AMR
- A range of educational materials and the use of different communication channels may be required to increase health literacy on the appropriate use of antibiotics in different age groups, and in the younger population specifically
- Healthcare professionals play a vital role in educating the public on appropriate antibiotic use. It is important that they understand the most prevalent misconceptions in their region as well as the population groups that require attention, so that messaging can be tailored accordingly

REFERENCES

1. World Health Organization. Antimicrobial resistance: Key facts. 2021. Available from: https://www.who.int/news-room/fact-sheets/detail/antimicrobial-resistance (accessed August 2023)
2. World Health Organization. https://apps.who.int/iris/bitstream/handle/10665/194460/9789241509817_eng.pdf (accessed August 2023)
3. Cals JW, et al. Br J Gen Pract 2007;57:942–7
4. Widayati A, et al. Antimicrob Resist Infect Control 2012;1:38
5. Gaarslev C, et al. Antimicrob Resist Infect Control 2016;5:39
6. Davis ME, et al. Antibiotics (Basel) 2017;6:23
7. World Health Organization. Global action plan on antimicrobial resistance. 2015. Available from: https://apps.who.int/iris/rest/bitstreams/864486/retrieve (accessed August 2023)
8. Altiner A, Duerden M, Kozlov R, Lopez-Pintor E, Sessa A, Shephard A, van der Velden AW. Understanding attitudes and perceptions of consumers in relation to antibiotic use for respiratory illness and symptoms in Europe. Presented at European Congress of Clinical Microbiology and Infectious Diseases (P2298), Copenhagen, Denmark, 15–18 April 2023

WEBSITE

www.grip-initiative.org

DISCLOSURES

This study was funded by Reckitt. AS is an employee of Reckitt. SE, SCB, AP, KE, WT and AvdV are members of the Global Respiratory Infection Partnership, which is supported by an unrestricted educational grant from Reckitt. Poster presented at the annual IDWeek meeting 2023, Boston, Massachusetts, 11–15 October 2023.

ACKNOWLEDGEMENTS

Medical writing support was provided by Honey McElhill, Elements Communications Ltd, UK, and was funded by Reckitt.